Modulation of dopamine D_{2L} and D_3 signalling and trafficking and cytosolic cell surface trafficking by dysbindin

N. Schmieg[1], C. Rocchi[2], R. Maggio[2], M. J. Millan[1] and C. Mannoury la Cour[1]

[1]Unite Recherche et Decouverte Neuroscience, Institut de Recherches Servier, Croissy-sur-Seine, France ;

[2]Biotechnological and Applied Clinical Sciences Department, University of L'Aquila, L'Aquila, Italy

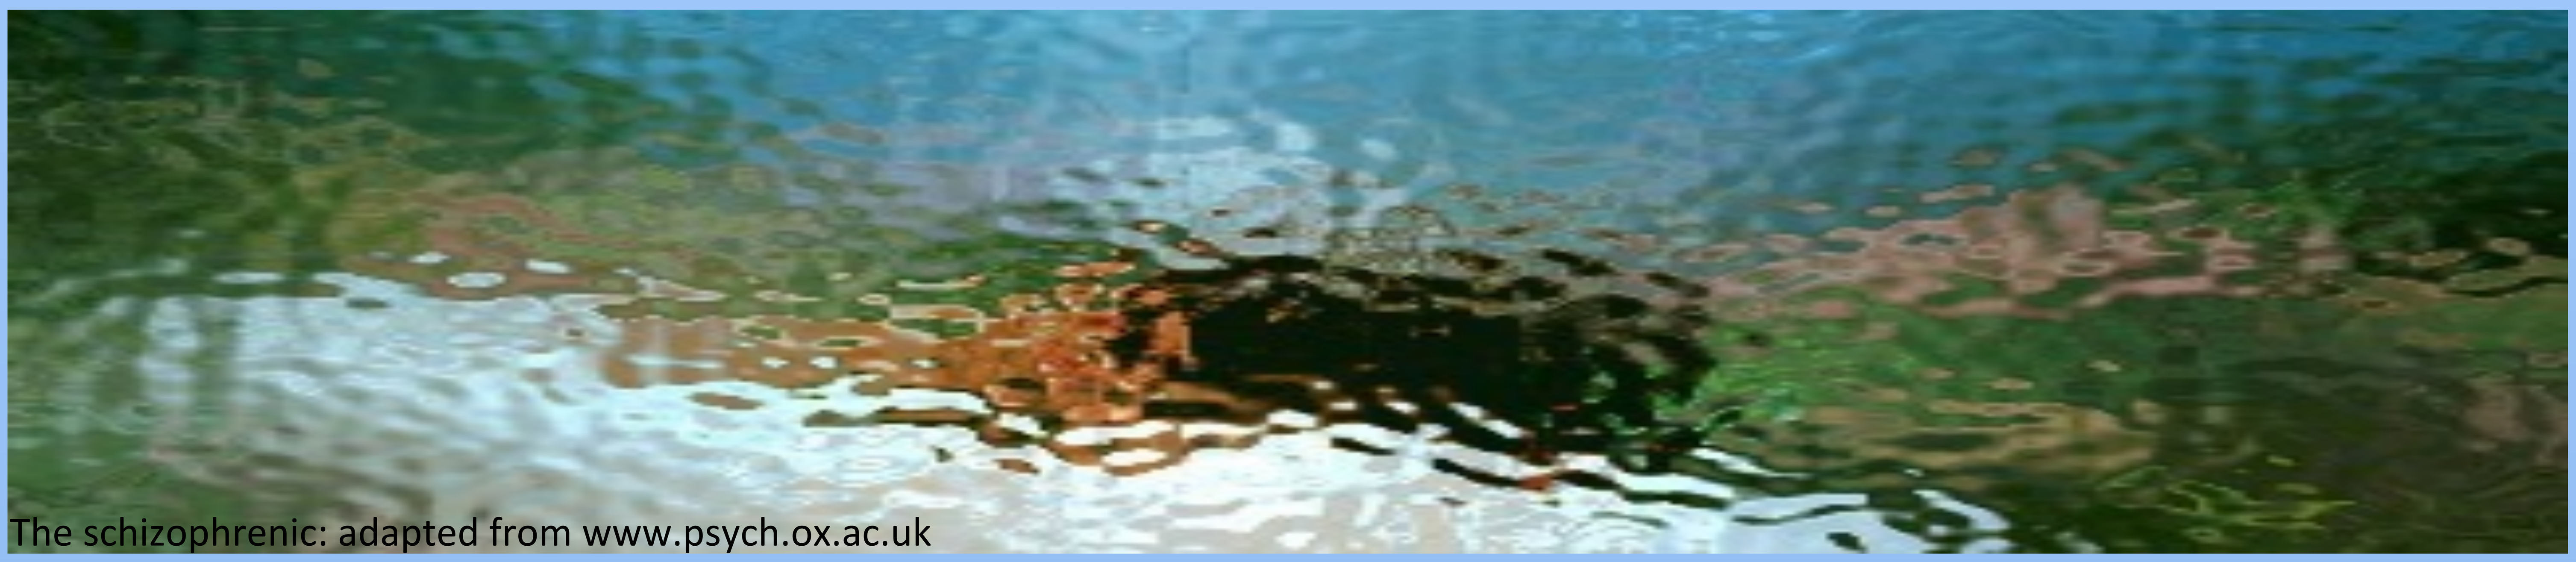

The schizophrenic: adapted from www.psych.ox.ac.uk

Abstract: Schizophrenia is a severe neuropsychiatric disorder with many risk factors, both environmental and genetic. In regards to genetic factors, dystobrevin binding protein 1 (dysbindin) is a susceptibility gene and its levels are reduced in schizophrenia [1]. Further, genetically reducing dysbindin expression enhanced cell surface expression of D_2 receptors in frontal cortex [2]. These observations suggest altered control of dopaminergic transmission in schizophrenia by dysbindin. In view of this we investigated the influence of dysbindin overexpression on signalling and cell surface localisation of human (h) dopamine D_{2L} versus D_3 receptors stably expressed in CHO cells.

I. Dysbindin decreases cell surface expression of Dopamine D_{2L} and D_3 receptors in CHO cells

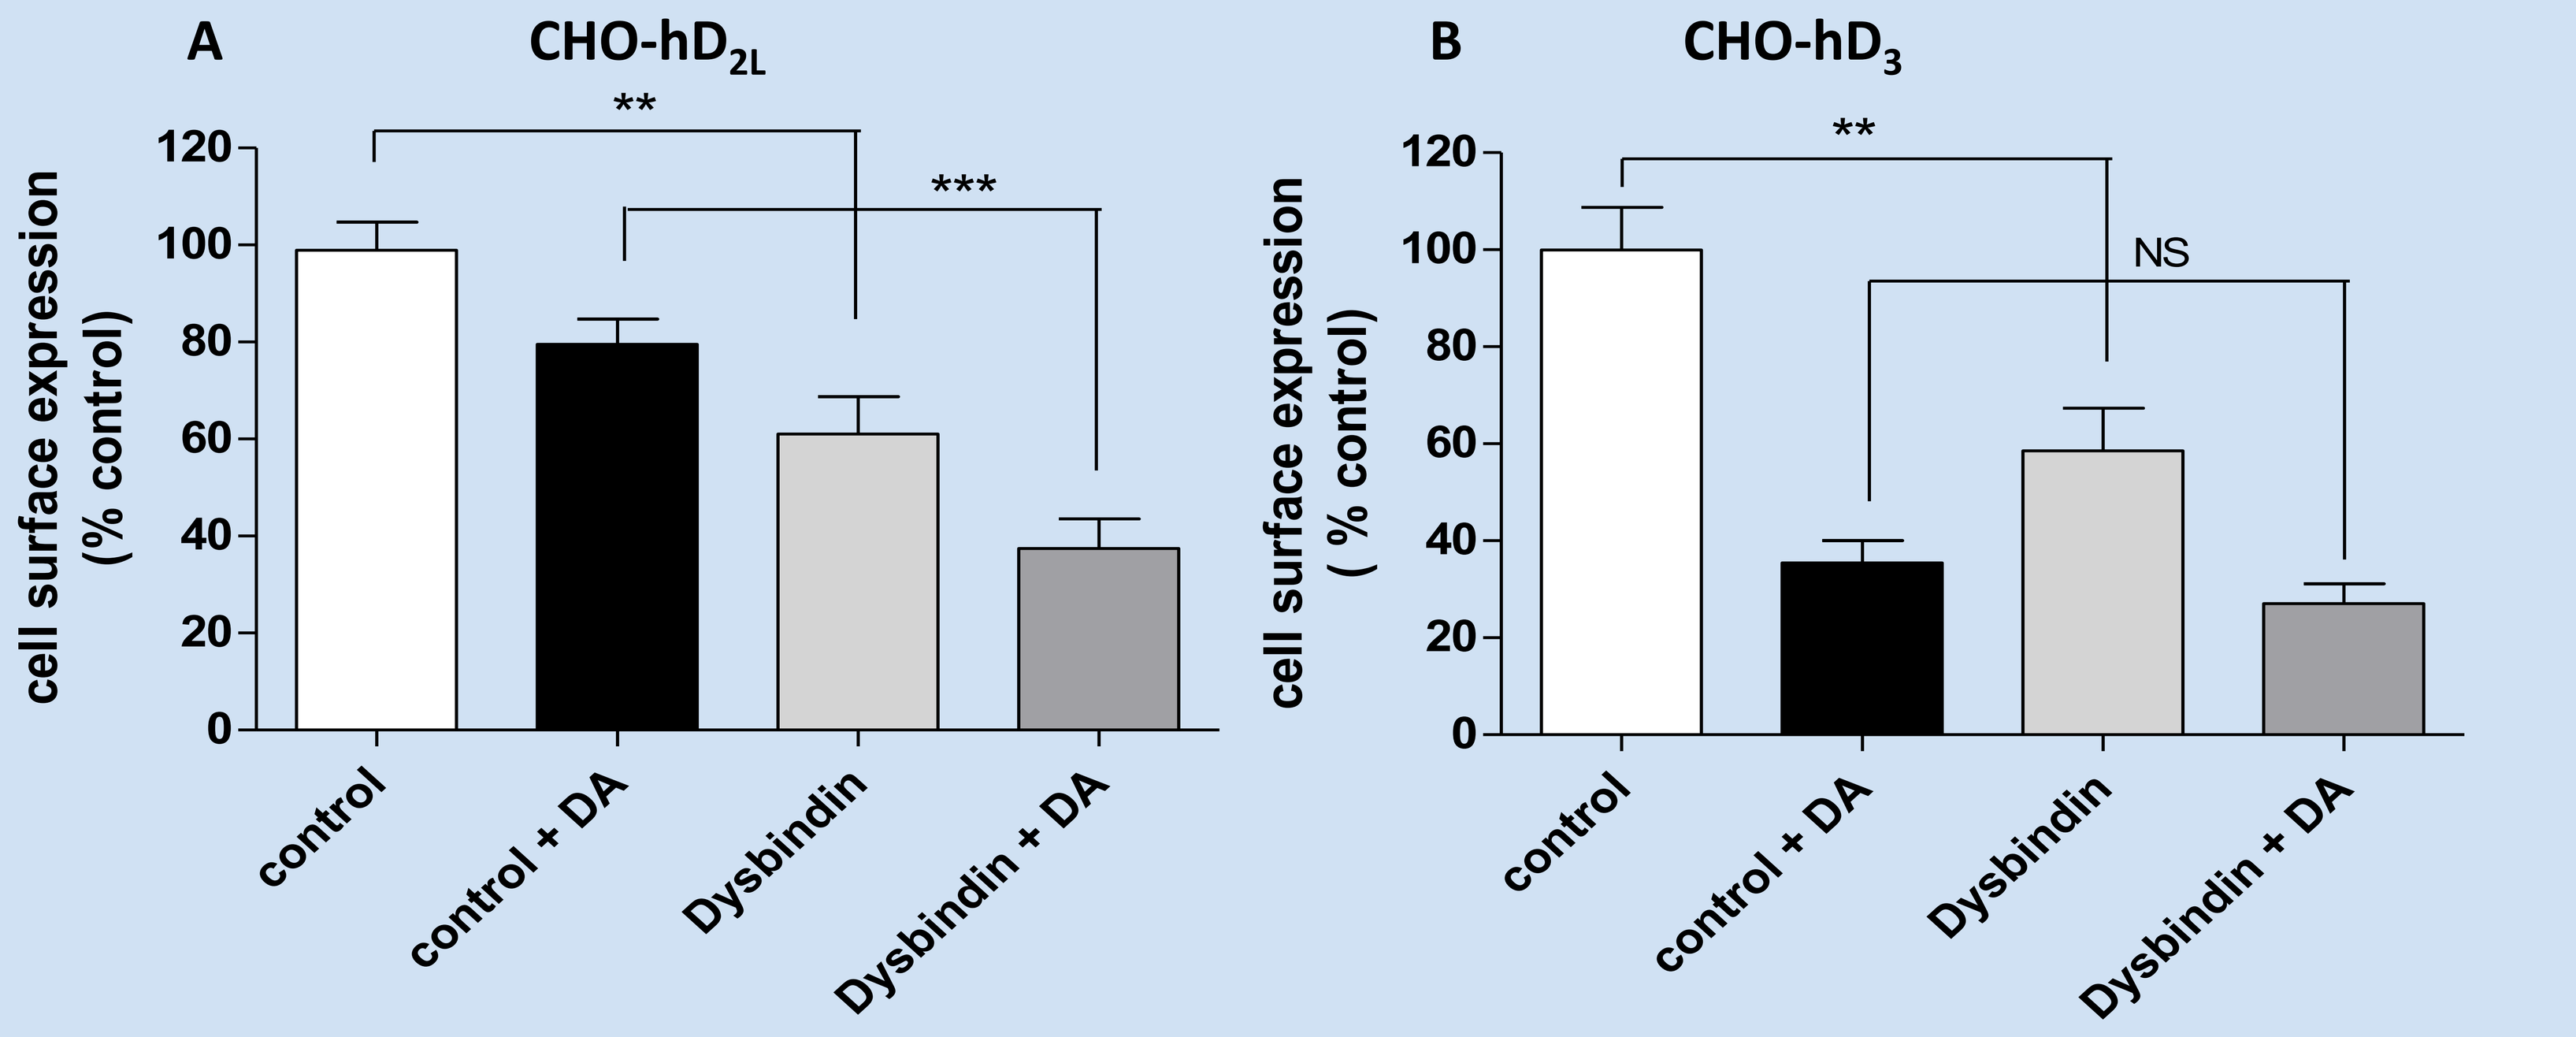
A
CHO-hD2L
B
CHO-hD3
cell surface expression (% control)
control
control + DA
Dysbindin
Dysbindin + DA
NS

CHO-hD_{2L} (A) and CHOhD_3 (B) were transiently transfected with dysbindin. Employing the radioligand [^{3}H]-methoxysulpride, dysbindin reduces the density of hD_{2L} and hD_3 receptors to 61±5 and 59±4%, respectively compared to control cells (100%), without any change in radioligand affinity (KD): 0.22±0.05 and 0.23±0.05, for hD_{2L} and hD_3 receptors, respectively (data not shown); Dopamine= DA.

II. Dysbindin decreases inhibitory effect of dopamine stimulation on forskolin-activated adenylyl cyclase

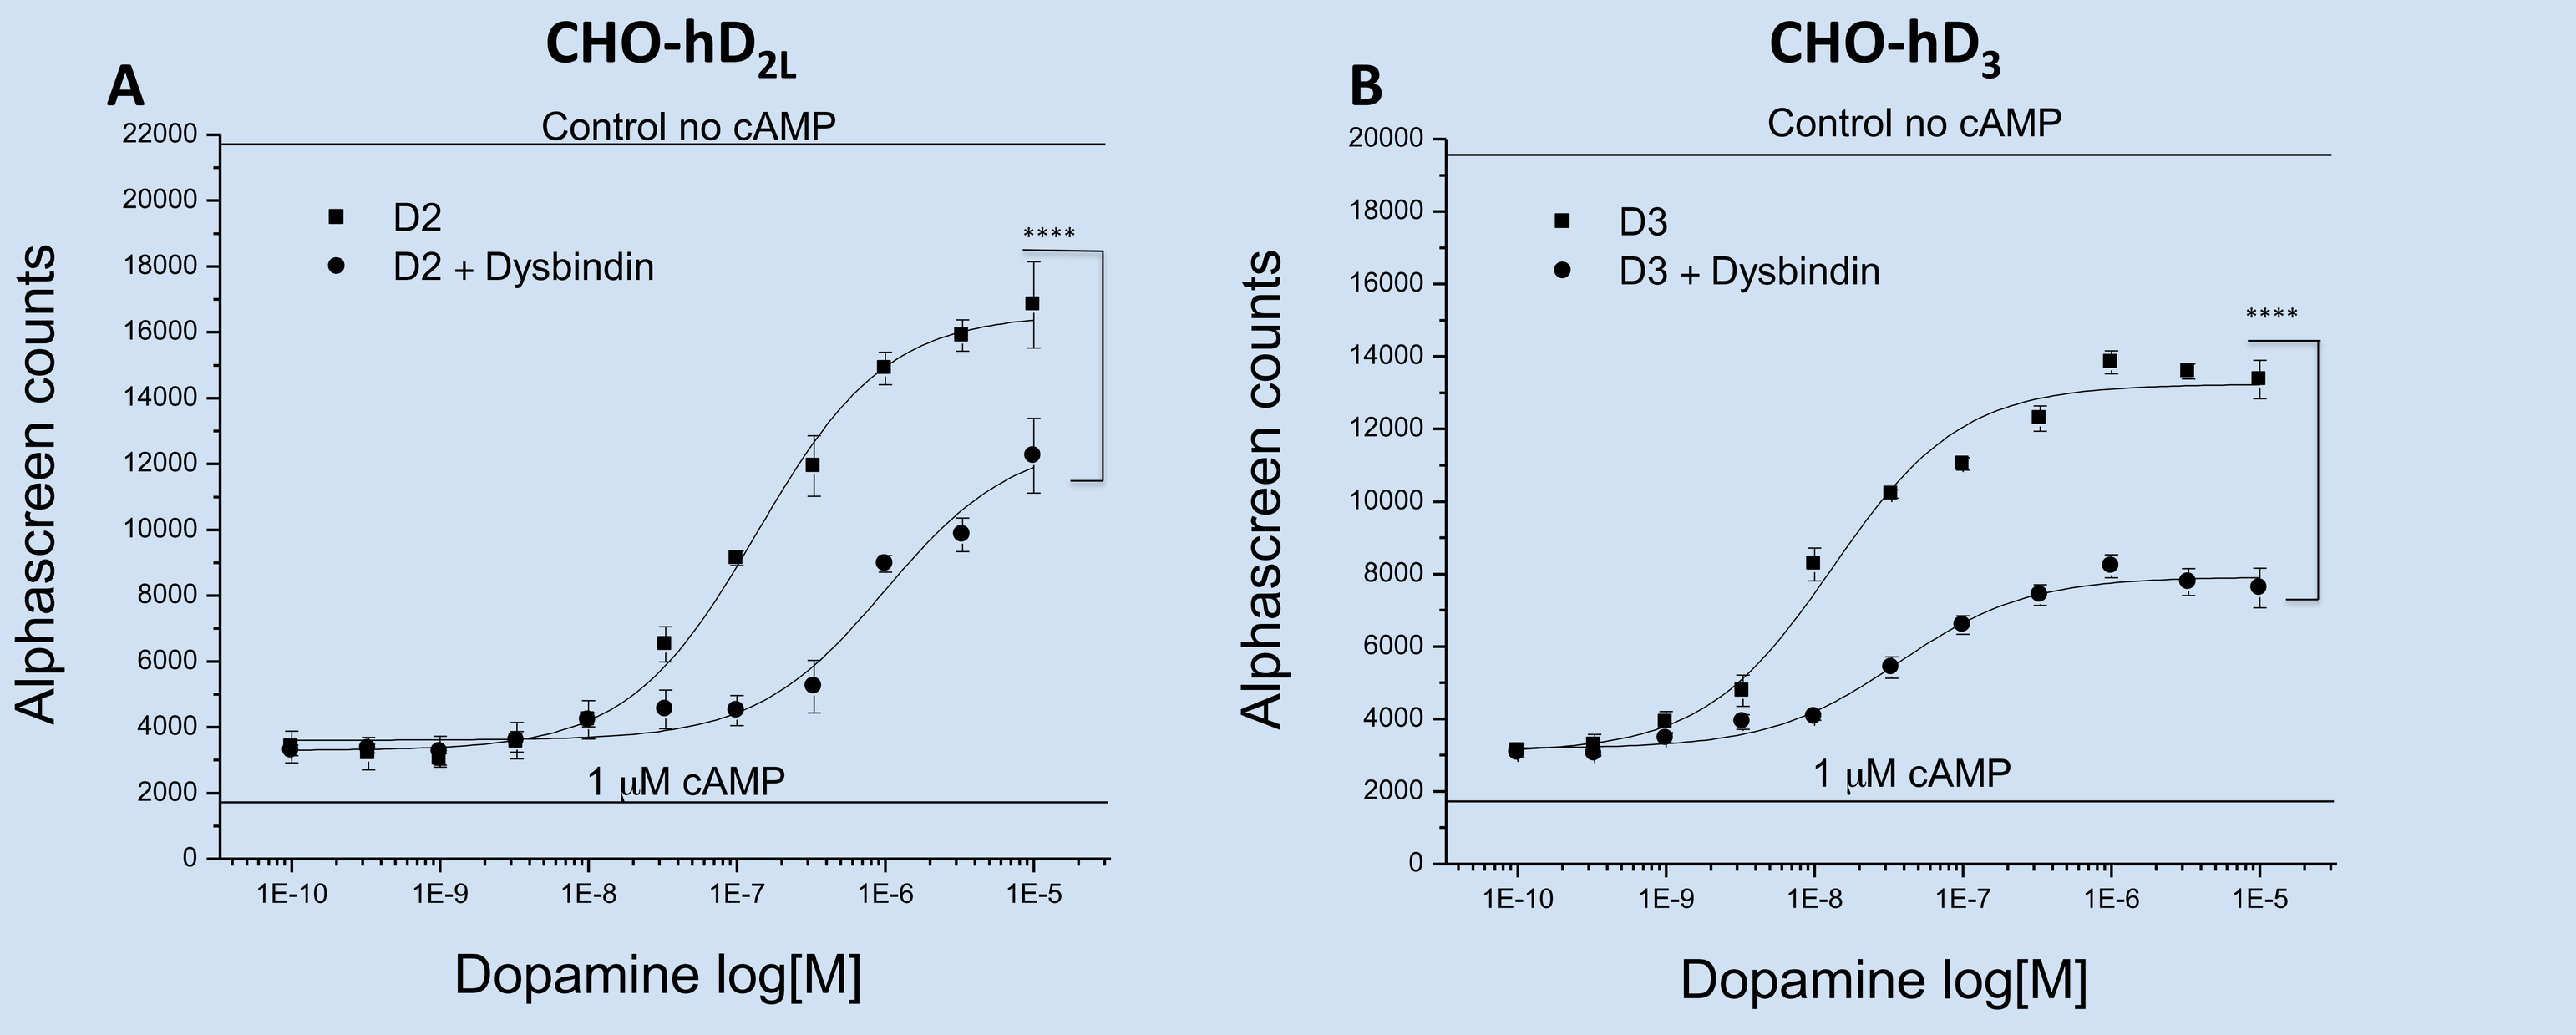
A
CHO-hD2L
Control no cAMP
D2
D2 + Dysbindin
1 μM cAMP
B
CHO-hD3
D3
D3 + Dysbindin
Alphascreen counts
Dopamine log[M]

Dysbindin overexpression decreases the inhibitory effect of DA stimulation on forskolin-activated adenylyl cyclase activity shown by this Alphascreen Surefire™ assay. As compared to control values (defined as 100%), values in the presence of dysbindin were 62±7% and 41±5% for hD_{2L} (A) and hD_3 (B) receptors, respectively –as compared to 98±6% for D_1 receptors, which were not affected in a control study(data not shown).

III. Dysbindin decreases efficacy of Akt and GSK-3β phosphorylation upon dopamine stimulation

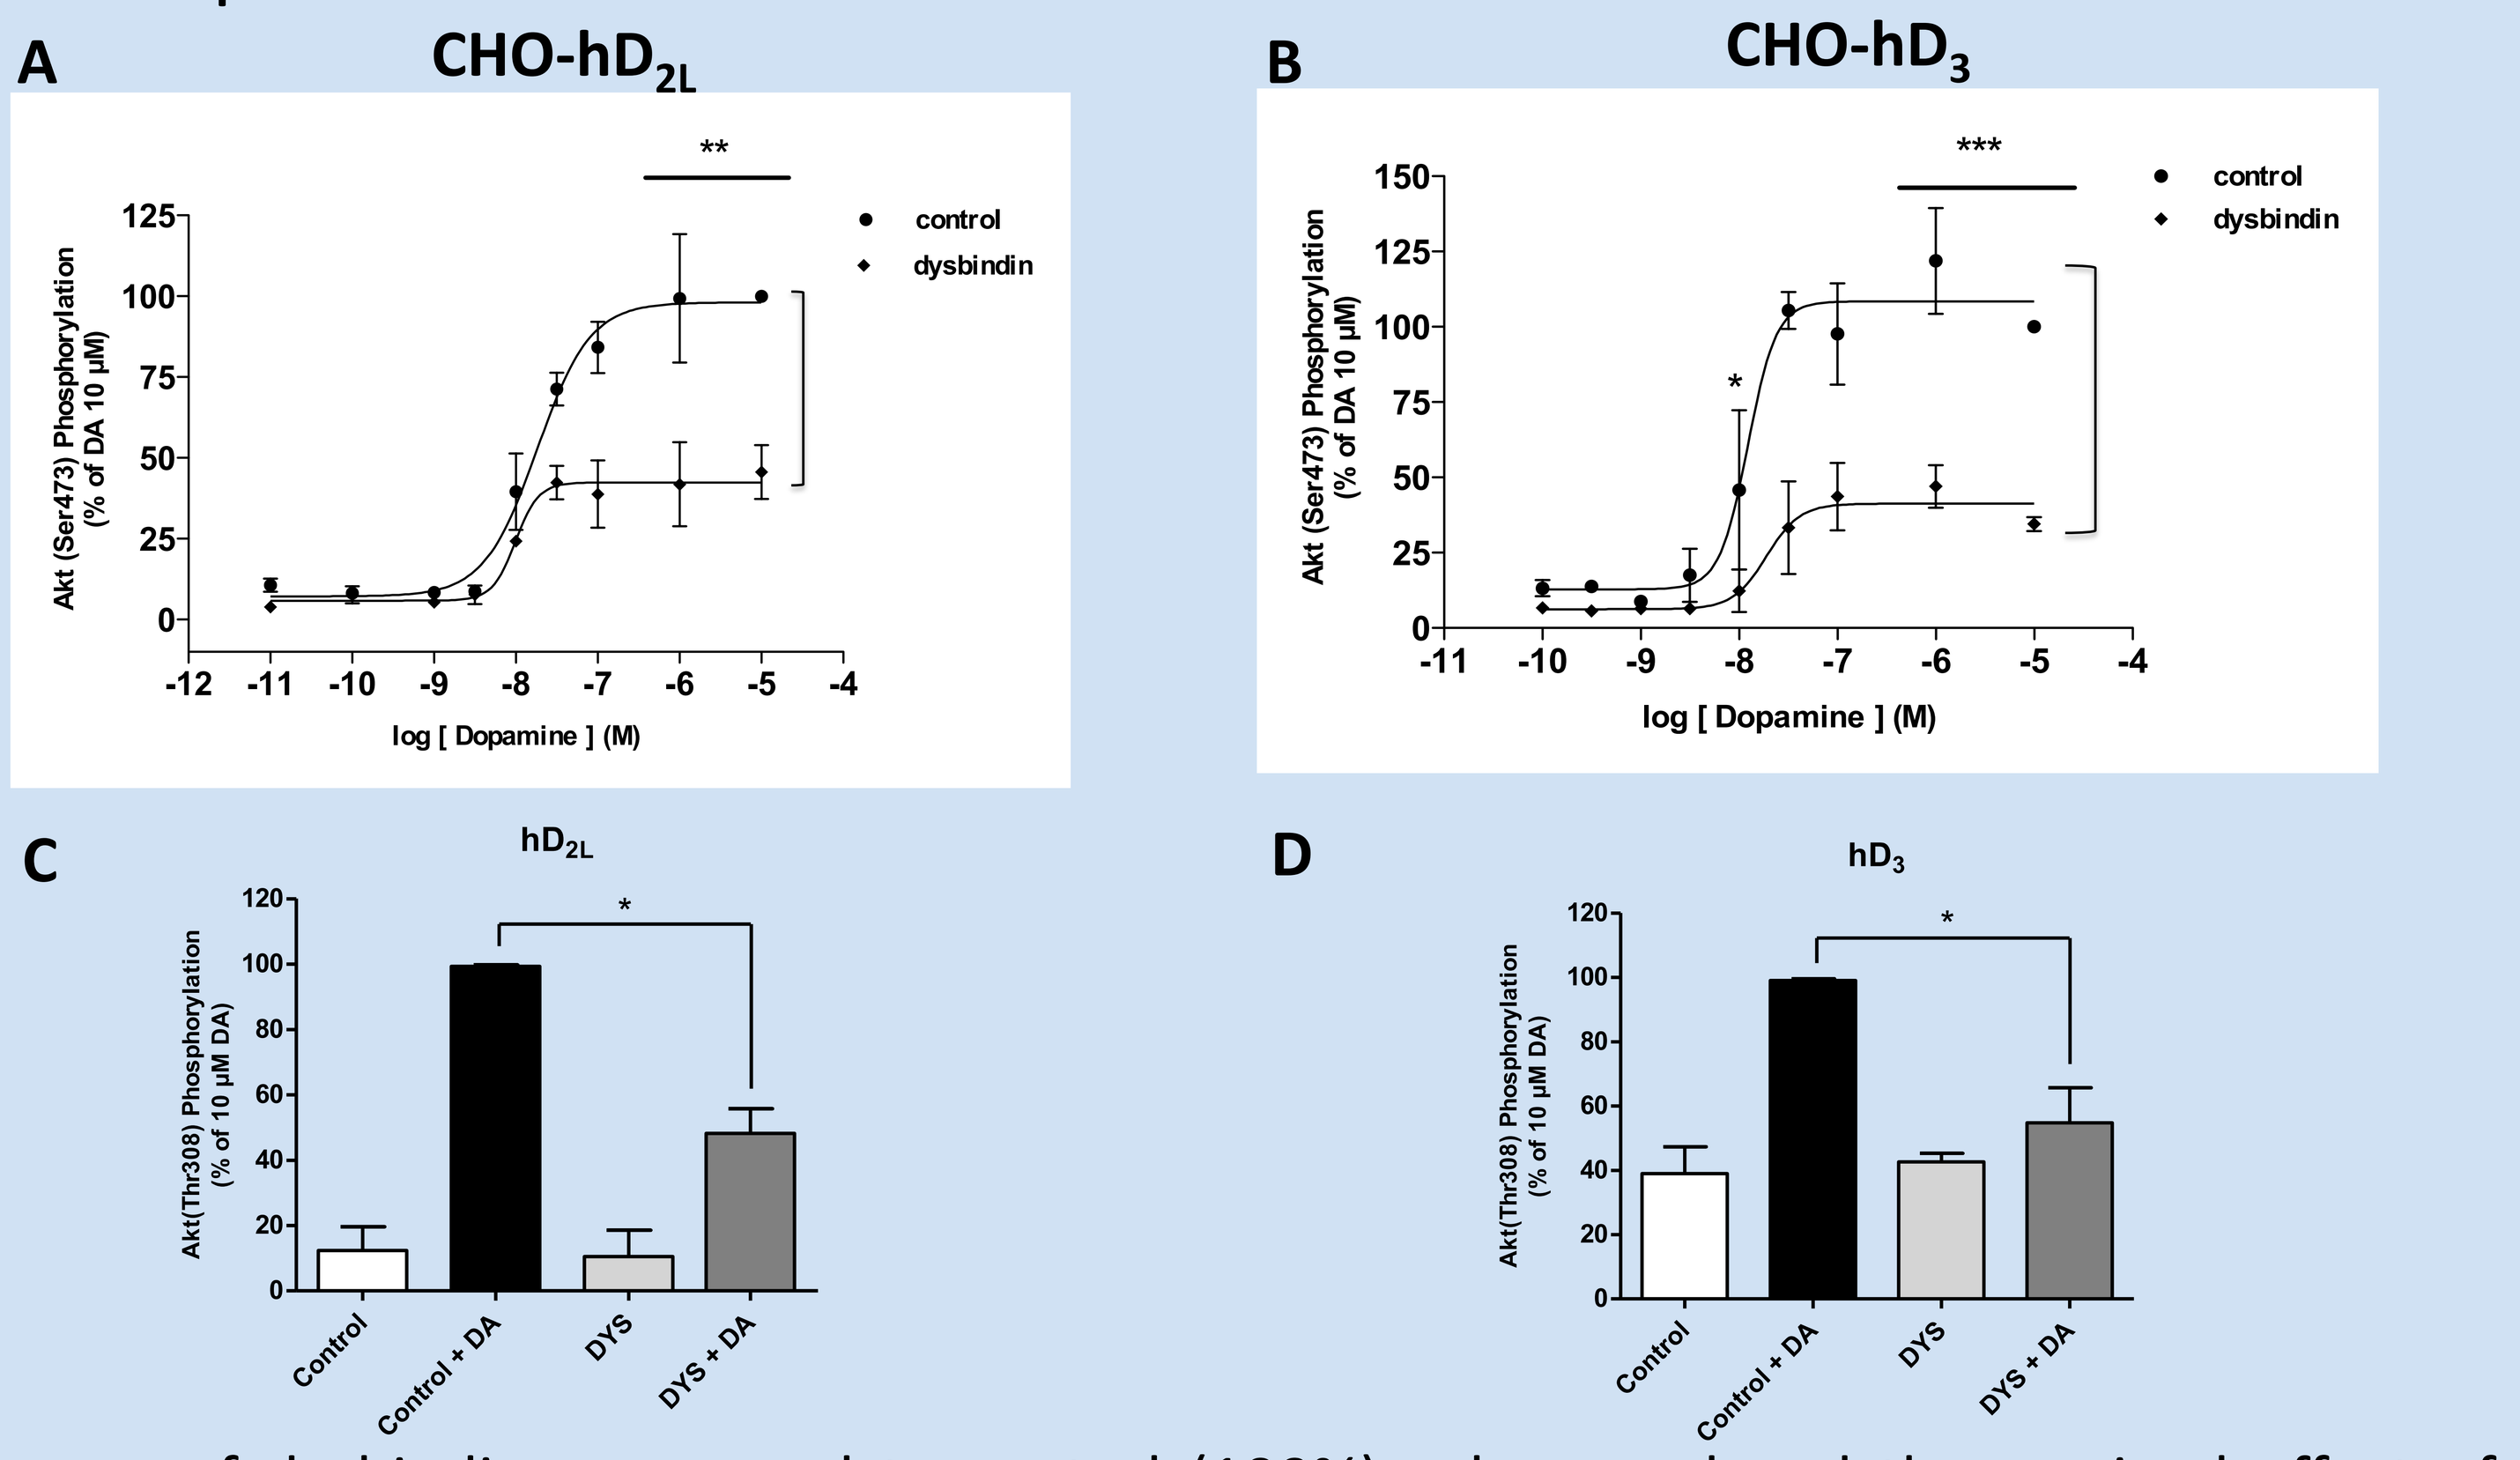
A
CHO-hD2L
B
CHO-hD3
control
dysbindin
Akt (Ser473) Phosphorylation (% of DA 10 μM)
log [Dopamine] (M)
C
hD2L
D
hD3
Akt(Thr308) Phosphorylation (% of 10 μM DA)
Control
Control + DA
DYS
DYS + DA

Presence of dysbindin -compared to control (100%) values- reduced the maximal effect of Akt(Ser473 and Thr308) and GSK-3β phosphorylation to 44±3 (A), 48±12(C) and 58±5% (figure not shown), respectively in CHO-hD_{2L} cells; In CHO-hD_3 these values were reduced to 41±4 (B), 45±19(D) and 53±3% (figure not shown), Data represent quantifications from three independent Western Immunoblots.

IV. Efficacy of Erk1/2 phosphorylation is decreased by dysbindin overexpression

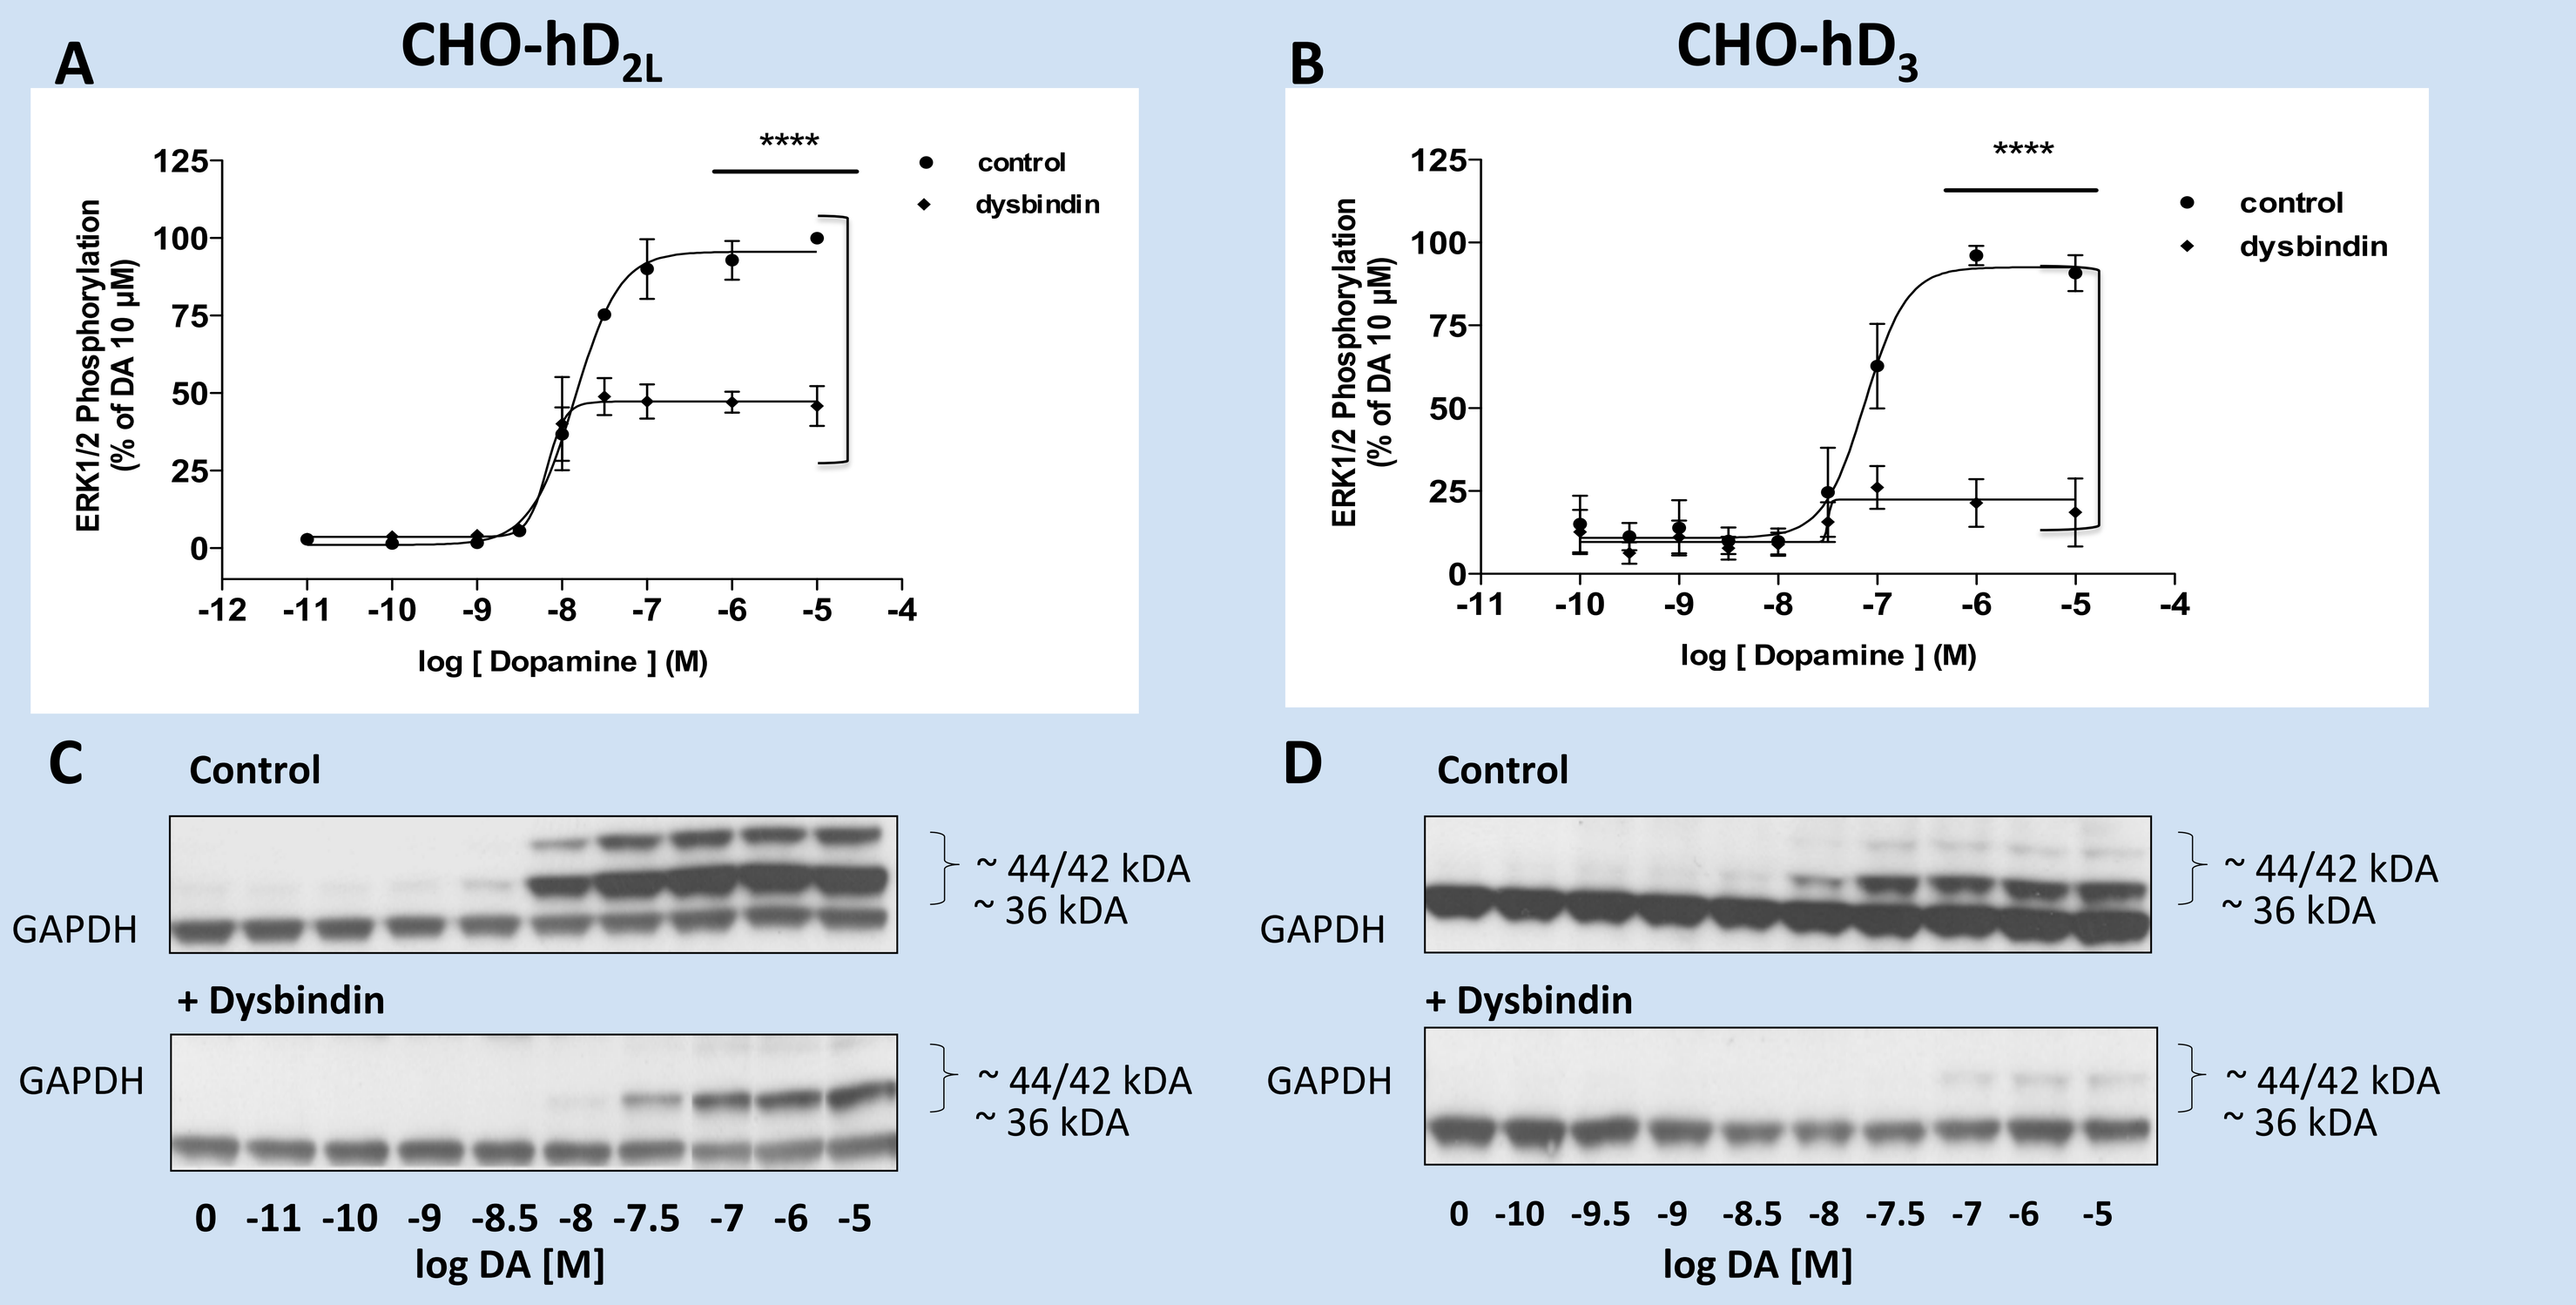
A
CHO-hD2L
B
CHO-hD3
control
dysbindin
ERK1/2 Phosphorylation (% of DA 10 μM)
log [Dopamine] (M)
C
D
Control
+ Dysbindin
GAPDH
~ 44/42 kDA
~ 36 kDA
0 -11 -10 -9 -8.5 -8 -7.5 -7 -6 -5
0 -10 -9.5 -9 -8.5 -8 -7.5 -7 -6 -5
log DA [M]

Presence of dysbindin compared to control (100%) values reduced the maximal effect of Erk1/2 phosphorylation to 48±3% in CHO-hD_{2L} cells (A&C) and by 22±4% in CHO-hD_3 cells (B&D). Data represent means of three independent Western Immunoblots (A&B). C&D are representative blots of A&B ,respectively.

V. Inhibition of clathrin/caveolar- mediated receptor internalisation attenuates the decreasing effect of dysbindin on hD_{2L} and hD_3 signalling

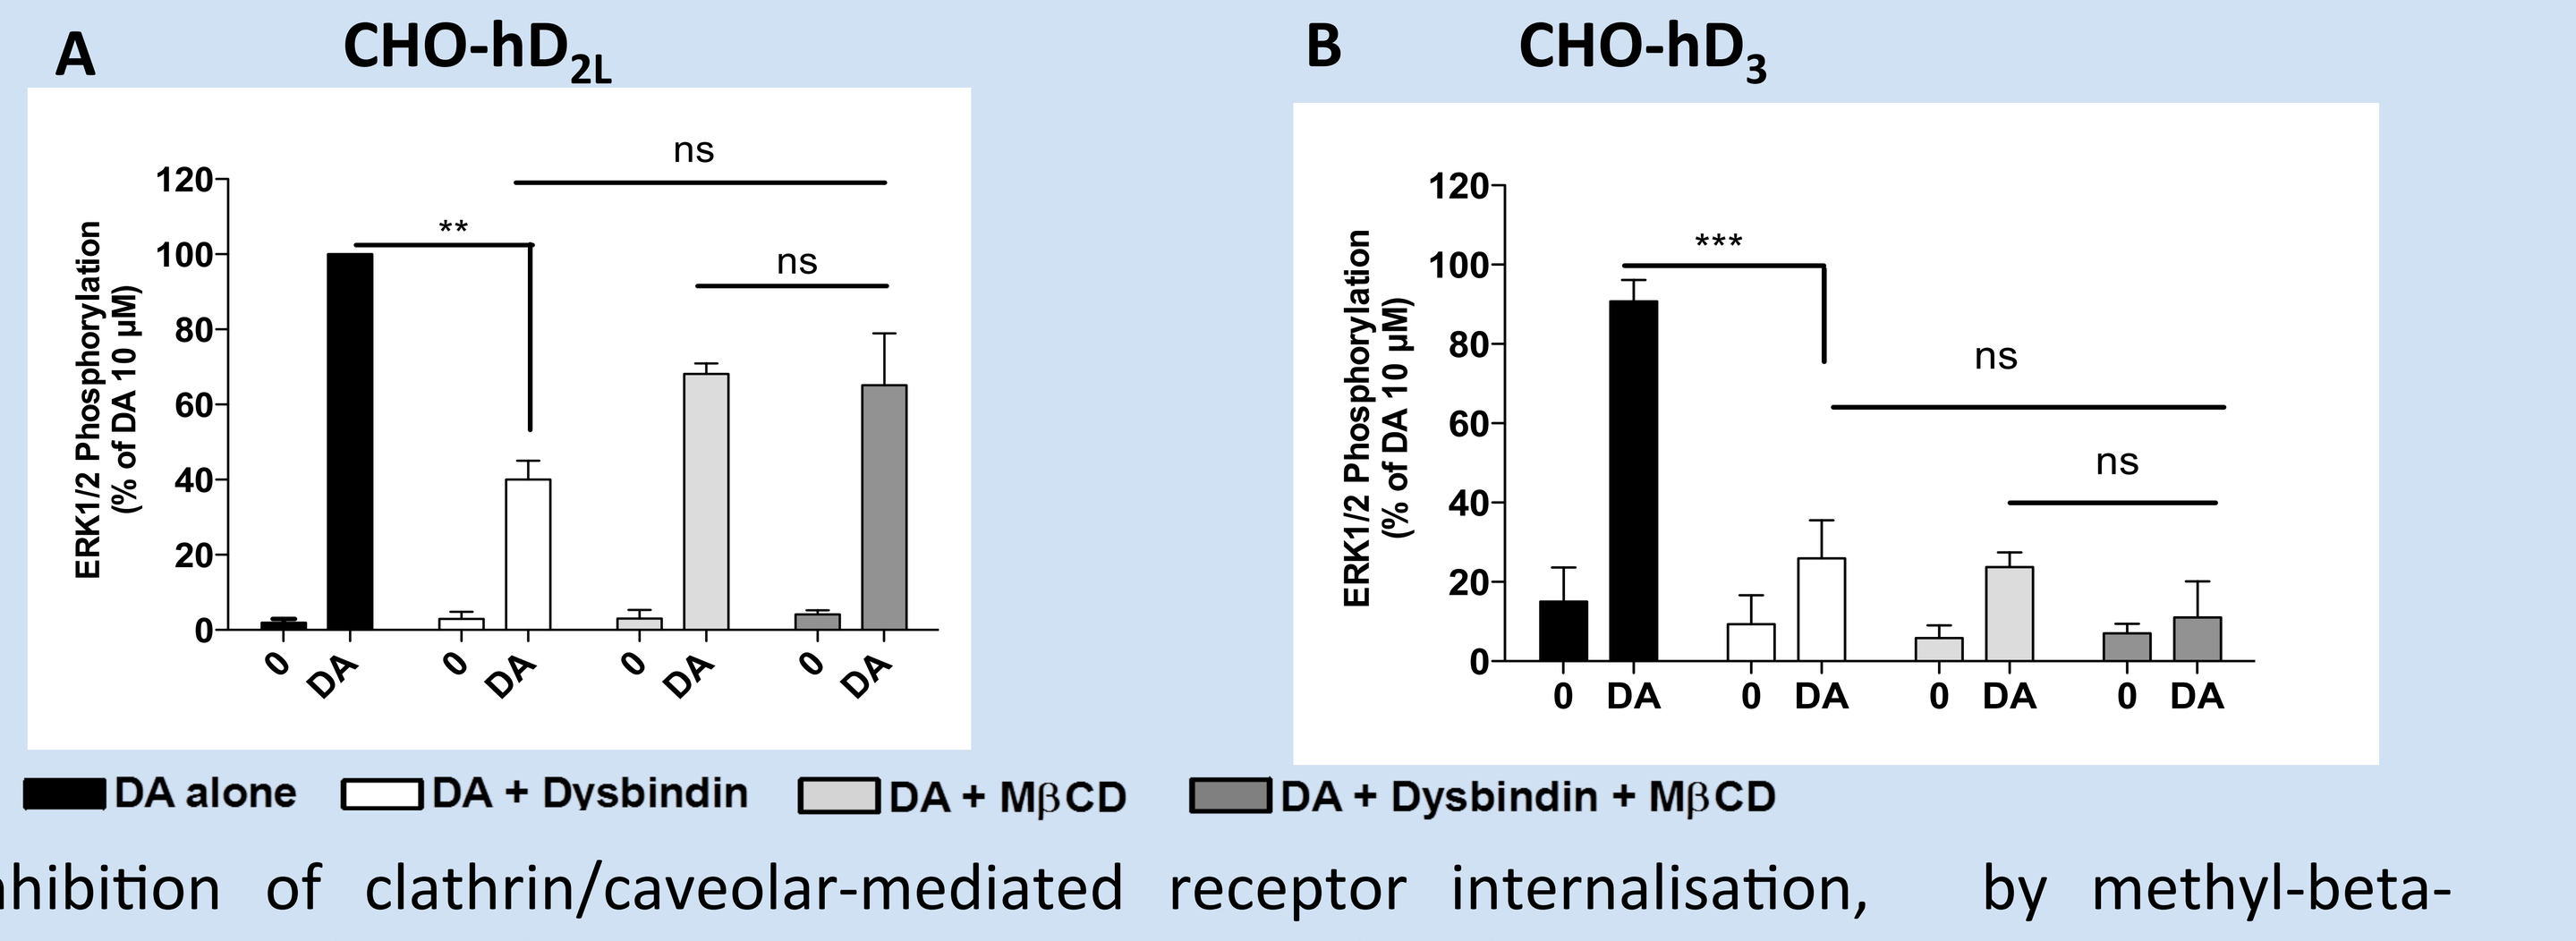
A
CHO-hD2L
B
CHO-hD3
ERK1/2 Phosphorylation (% of DA 10 μM)
ns
DA alone
DA + Dysbindin
DA + MβCD
DA + Dysbindin + MβCD

Inhibition of clathrin/caveolar-mediated receptor internalisation, by methyl-beta-cyclodextrin (MβCD), caused a slight reduction (32%) of the DA-mediated increase in ERK1/2 phosphorylation and completely prevented the effect of dysbindin overexpression (35%) CHO-hD_{2L} cells. However, in CHO-hD_3 cells (B) MβCD largely attenuated the effect of DA-induced increase of pERK1/2 phosphorylation (76%) but prevented further inhibition due to the overexpression of dysbindin (89%).

VI. Conclusions

Overexpression of dysbindin:

- reduces the efficacy of DA-mediated signalling of hD_{2L} and hD_3 receptors
- reduces inhibition of the adenylyl cyclase and recruitment of Akt/GSK-3β and ERK1/2 pathways in both receptor signalling pathways (hD_{2L} and hD_3).
- decreases cell surface expression of hD_{2L} and hD_3 receptors and may reflect a decrease of receptor density involving clathrin/caveolar-mediated internalisation.

Abnormalities in dysbindin control of D_3 and D_{2L} receptor signalling and localization may be related to the pathogenesis and symtpoms of psychosis.

VII. References

[1] Ghiani C.A. and Dell'Angelica E.C., 2011. Dysbindin-containing complexes and their proposed functions in the brain: From zero to (too) many in a decade. ASN neuro 3. pii: e00058.

[2] Ji, Y, Yang, F., Papaleo, H.X., Wang, W.J. Gao, D.R. Weinberger, D.R. et B. Lu., 2009. Role of dysbindin in dopamine receptor trafficking and cortical GABA function. Proc Natl Acad. Sci. 106, 19593-19598.

VIII. Disclosure

This work has been supported by the Unite Recherche et Decouverte Neuroscience, Institut de Recherches Servier, Croissy-sur-Seine, France